FINAL RESULTS OF ANAGRELIDE CONTROLLED-RELEASE (GALE-401) SAFETY, EFFICACY AND PHARMACOKINETICS IN SUBJECTS WITH MYELOPROLIFERATIVE NEOPLASMS (MPN)-RELATED THROMBOCYTOSIS

Truong P[1], Saltzman M[2], Bessudo A[3], Jawien W[4], Lyons R[5], Berenzon D[6], Mena R[7], Wingate-Pearse N[8], Barriere O[9], Beeson H[10], Glidden P[8], Choy GS[8], Verstovsek S[11]

[1]Cancer Center of Kansas, Wichita, KS, [2]Innovative Medical Research of South Florida, FL, [3]California Cancer Associates for Research and Excellence, Encinitas, CA, [4]California Cancer Associates for Research and Excellence, Fresno, CA, [5]Cancer Care Centers of South Texas/USONcology, San Antonio, TX, [6]Wake Forest University School of Medicine, Winston-Salem, NC, [7]East Valley Hematology and Oncology Medical Group, Burbank, [8]Galena Biopharma, Inc., Portland, OR, [9]Inventiv Health Clinique, Inc., Montreal, Canada, [10]Chiltern International, Inc., Wilmington, NC, [11]University of Texas, MD Anderson Cancer Center, Houston, TX.

Background

Anagrelide (Agrylin® (United States) and Xagrid® (European Union)) is an immediate release (IR) agent that blocks megakaryocyte differentiation and proliferation and inhibits the action of cyclic AMP phosphodiesterase. Anagrelide hydrochloride is extensively metabolized to 2 major metabolites: 3-hydroxy-anagrelide (3-OH) and a subsequent biotransformation product, RL603. The metabolite 3-hydroxy-anagrelide is equipotent with the parent drug in its *in vitro* effects on megakaryocytopoesis and is therefore potentially platelet-lowering, and published studies have identified 3-hydroxy-anagrelide as active (Ahluwalia M et al. 2010). Anagrelide was developed as an inhibitor of platelet aggregation but was later found to reduce platelet count at doses lower than the amount needed for platelet aggregation. An alternate formulation that modifies this pharmacokinetic (PK) profile may improve patient tolerability and treatment outcomes. This led to the development of a controlled-release (CR) formulation of anagrelide (GALE-401). Ninety-eight healthy volunteer (HV) subjects have been enrolled among five Phase 1 clinical trials of GALE-401, demonstrating platelet lowering activity and favorable PK characteristics (Laliberte RJ et al., ASH 2014). These data provided support of the ongoing Phase 2 (GALE-401-201) study in subjects with MPN-related thrombocytosis, previously reported by Vertovsek S et al., EHA 2015). This report presents at least nine months of study treatment for all subjects active on trial, safety and pharmacokinetics updated from the last published report.

STUDY GALE-401-201

◆ **Introduction:**

- GALE-401-201: Phase 2, pilot, single arm, open label, multi-center study evaluating the efficacy and safety of anagrelide controlled release (CR) in subjects with thrombocytosis secondary to essential thrombocythemia (ET) and other myeloproliferative neoplasms (MPN); ClinicalTrials.gov Identifier: **NCT02125318.**

◆ **Objectives:**

- Primary objective: Estimate the overall platelet response rate (ORR).
- Secondary objectives: Safety, tolerability and pharmacokinetics (PK).

◆ **Major Eligibility:**

- Provide written informed consent.
- Male or female ≥ 18 years of age.
- Diagnosed with a MPN related elevated platelet count to include (chronic myelogenous leukemia [CML], polycythemia vera [PV)], primary myelofibrosis [PMF], or ET, based on the 2008 WHO classification of myeloid malignancies.
- Platelet count ≥ 600 10^9/L on two occasions at least 14 days apart prior to first dose of study drug.
- MPN diagnosis other than ET, concurrent anti-MPN treatment is permitted, provided that the doses are stable at least 4 weeks prior to first dose of study drug.
- Currently not receiving therapy specifically intended to reduce platelet counts.

◆ **Methods:**

- Anagrelide CR is administered at a starting dose of 0.5 mg twice daily (1.0 mg/day). The dose will be titrated at weekly intervals, on an individual basis, to determine the lowest dose required to achieve and maintain a target platelet count of 150–400 x 10^9/L.
 - Platelet response is defined as complete response (CR, ≤ 400 x 10^9/L) or partial response (PR, ≤ 600 x 10^9/L or ≥ 50% reduction from baseline) maintained for at least 4 weeks.
 - Toxicities based on NCI CTCAE v4.03.

Results

Table 1. Baseline Subject Characteristics

Characteristics	n	%
Sex		
Male / Female	8 / 10	44.0 / 56.0
Age (years)		
Mean / Median (range)	63 / 64 (40 – 79)	
MPN Diagnosis		
ET / PV	14 / 4	77.8 / 16.7
Mean Time in Weeks Since Diagnosis to Study Entry (range)	14 (1 – 55)	
Essential Thrombocythemia	65.8 (26.2 – 197.6)	
Polycythemia Vera	371.1 (61.7 – 758.3)	
Prior Treatments		
Other	3	16.7
Hydroxyurea	11	61.1
Anagrelide	8	44.4
Mutation		
JAK2 V167F / JAK2 Unknown	10 / 2	55.6 / 11.1
CALR / MPL	3 / 1	16.7 / 5.6
Mean Platelet Count x 10^9/L (range)	1046.9 (595 – 1620)	
Mean Hematocrit g/L (range)	41.2 (34 – 46)	
Aspirin / Clopidogrel (n)	12 / 1	72.0
Hypertension (n) / Diabetes mellitus (n)	13 / 1	72.0 / 5.6

Figure 1. Mean Platelet / Time

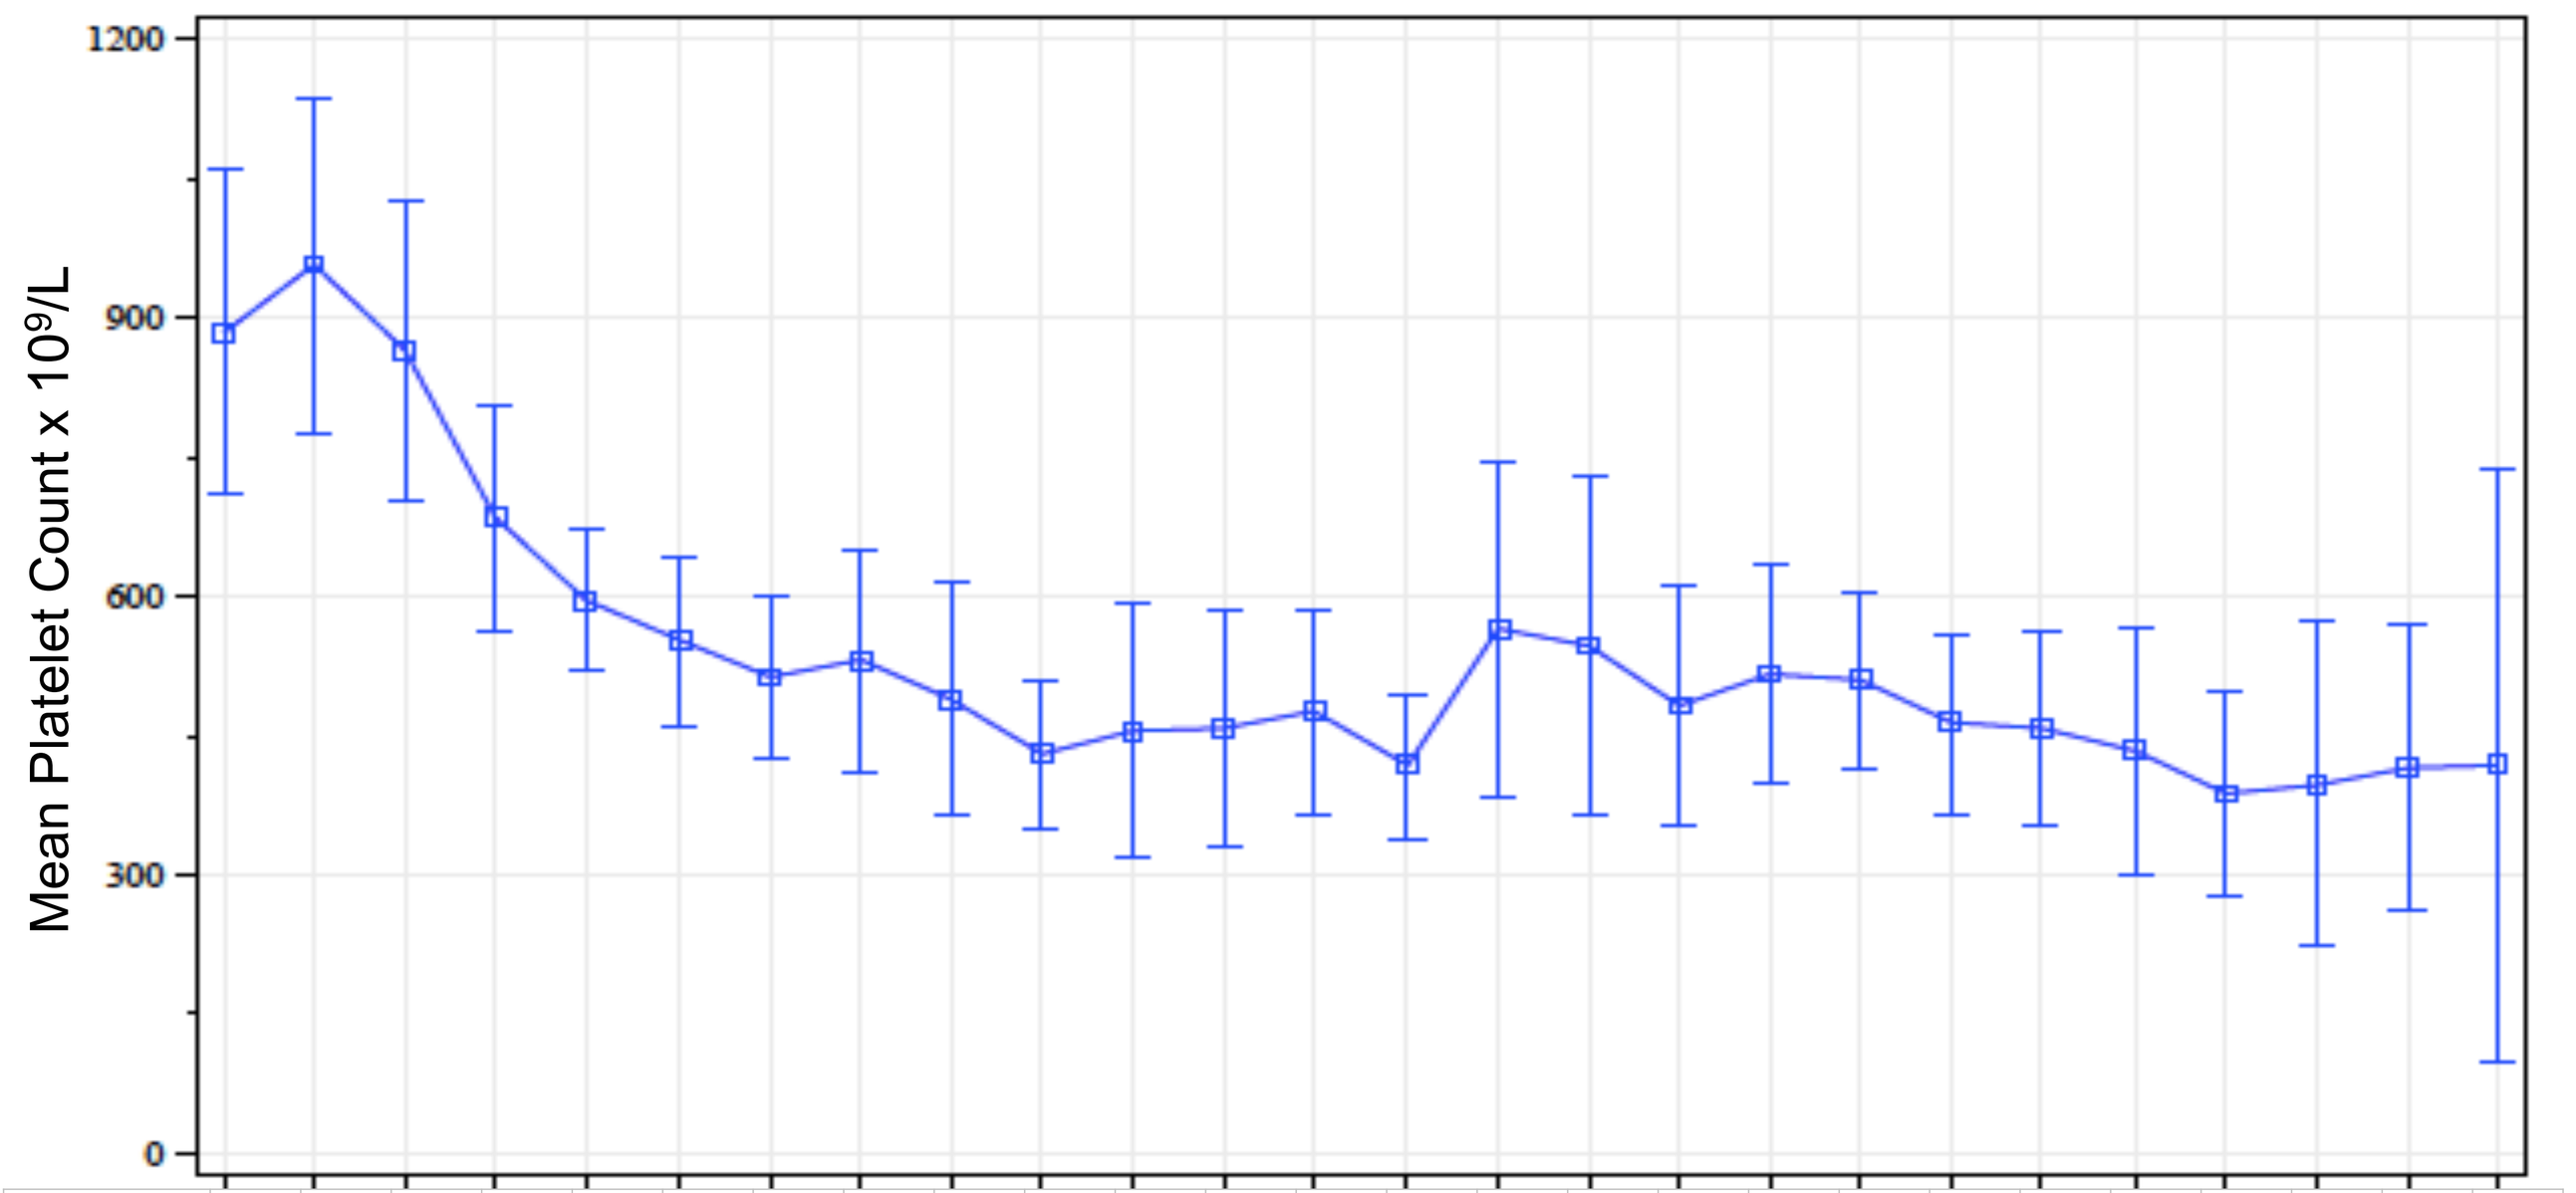
Mean Platelet Count x 10⁹/L
0
300
600
900
1200

Visit	SCRN	W1D1	W2D8	W3D15	W4D22	W5D29	W6D36	W7D43	W8D50	W9D57	W10D64	W11D71	W12D78	W13D85	W15D99	W17D113	W19D127	W21D141	W23D155	M7	M8	M9	M10	M11	M12	M14
Plt	885	1047	865	684	597	552	513	530	490	430	455	458	475	417	565	548	482	516	510	462	457	433	387	397	415	418
N=	18	18	18	18	18	18	18	18	15	14	14	12	12	12	12	13	13	12	12	11	11	10	9	8	7	5

Figure 2. Time to Response by Anagrelide Dose Group

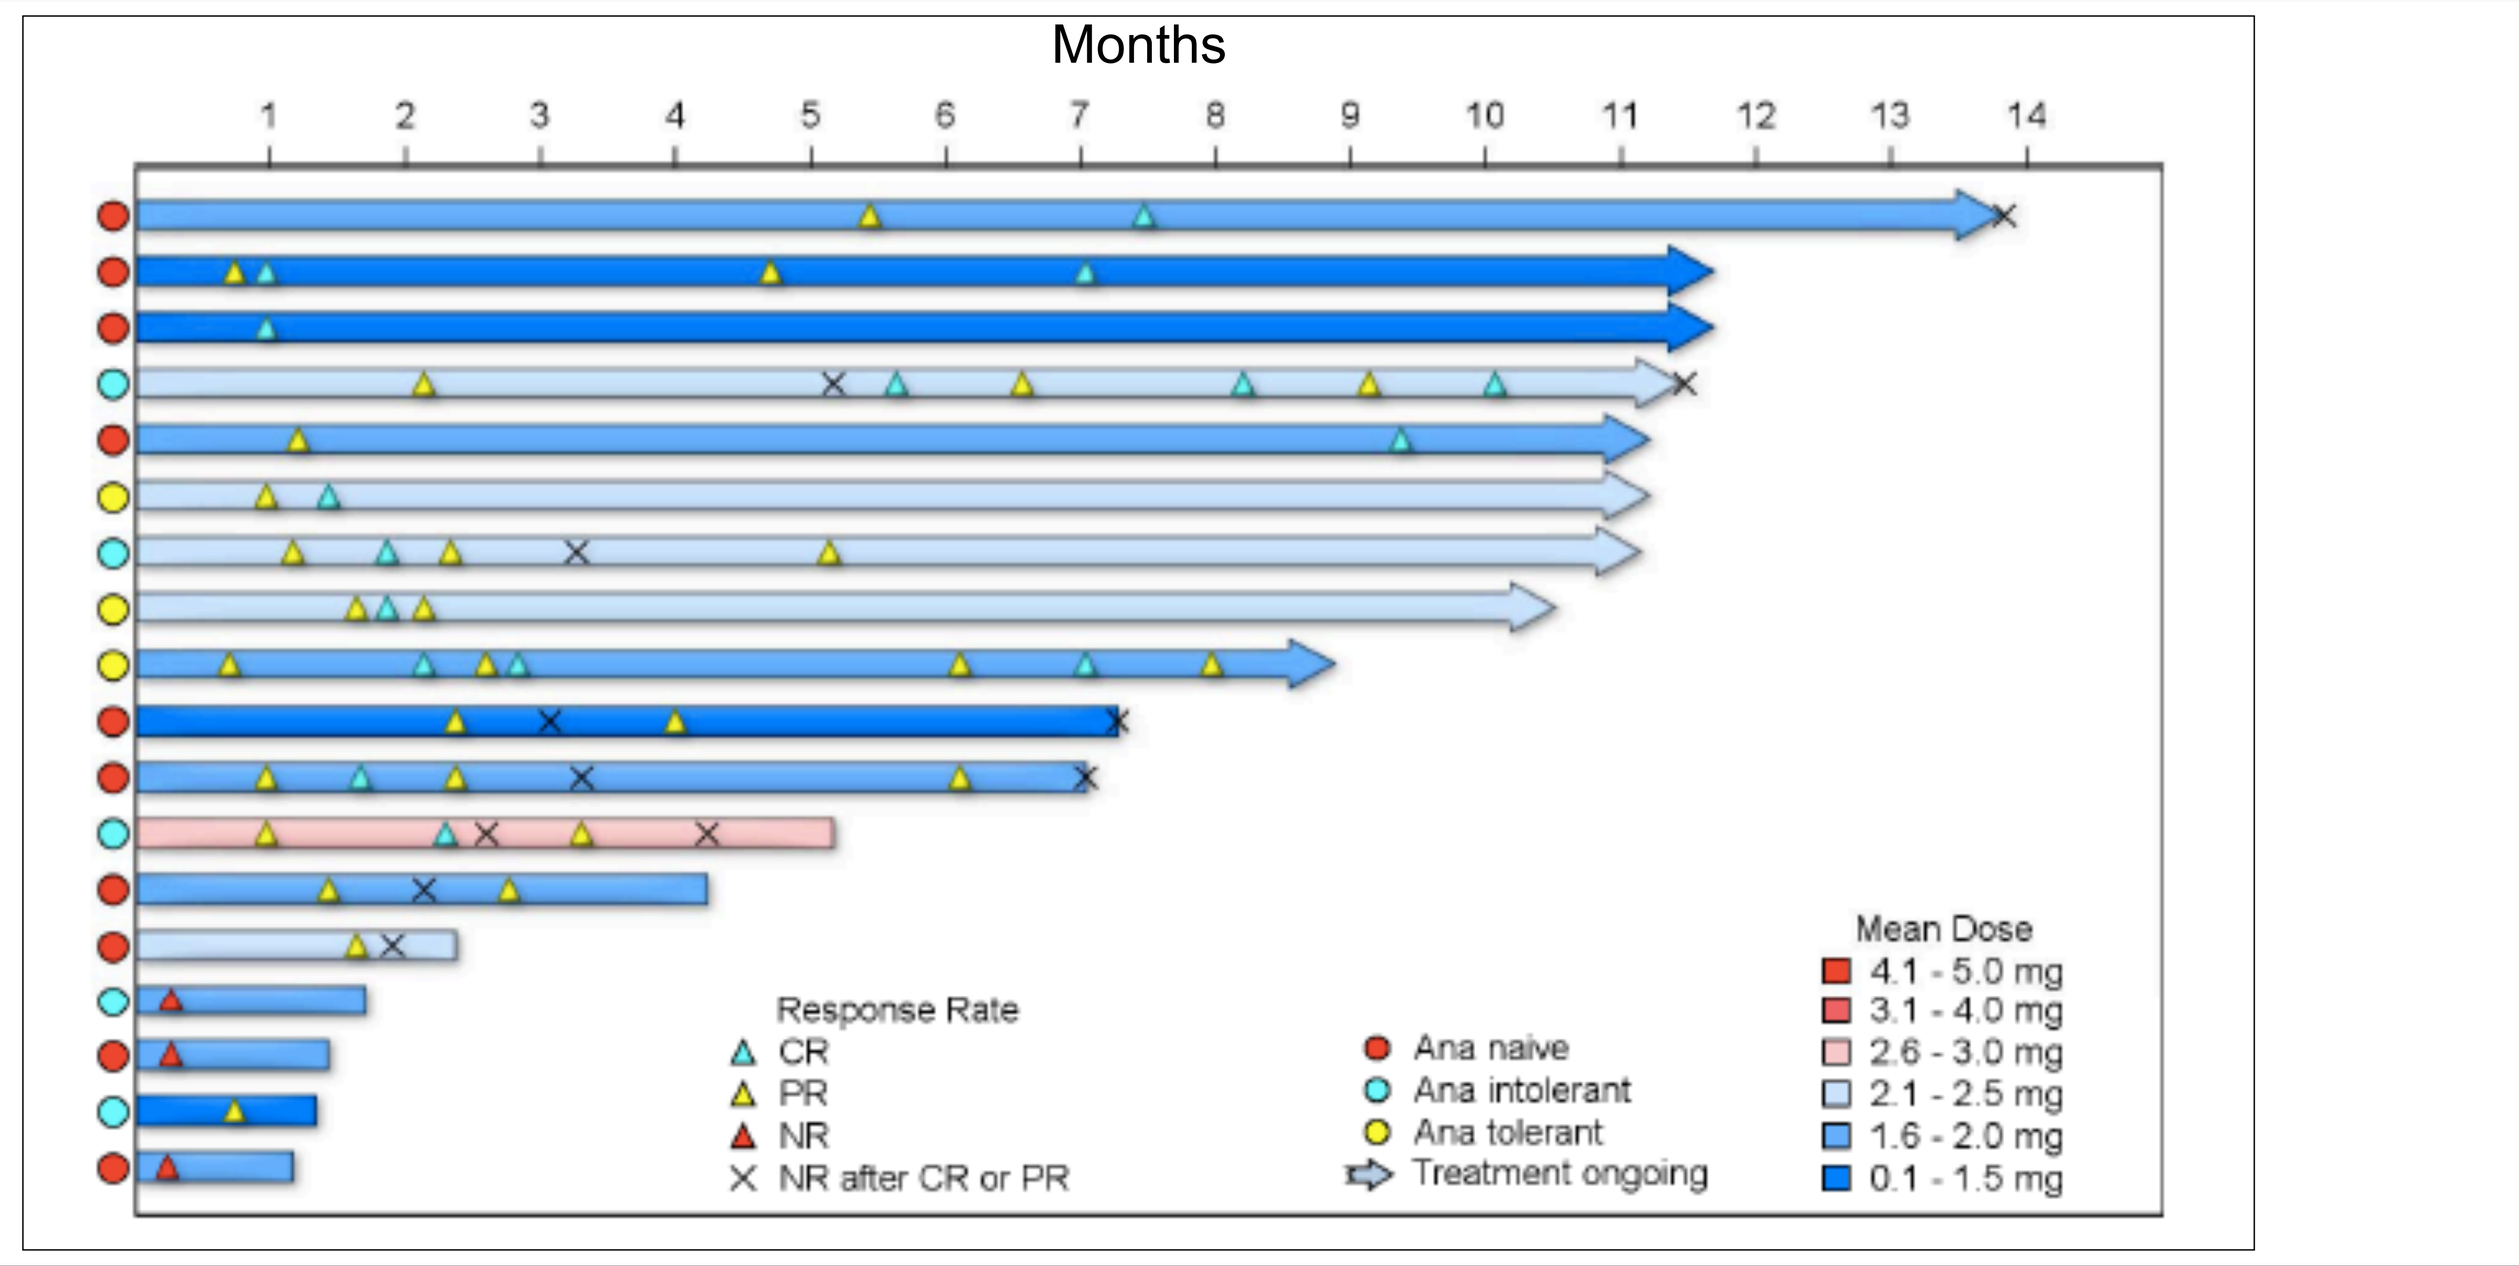
Months
1
2
3
4
5
6
7
8
9
10
11
12
13
14
Response Rate
CR
PR
NR
NR after CR or PR
Ana naive
Ana intolerant
Ana tolerant
Treatment ongoing
Mean Dose
4.1 - 5.0 mg
3.1 - 4.0 mg
2.6 - 3.0 mg
2.1 - 2.5 mg
1.6 - 2.0 mg
0.1 - 1.5 mg

- Time to mean platelet count <600 x 10^9/L is 23 days (range = 7 – 63) or 3.2 weeks (range = 1 – 9).
- Median daily dose over time = 2 mg.
- Nine MPN subjects remain active on study. Median duration of follow-up is ongoing.

Table 2. Best Response by Anagrelide Dose Group

Group	CR	PR	NR
Anagrelide IR Intolerant	3	1	1
Anagrelide IR Tolerant	3	0	0
Anagrelide IR Naive	5	3	2
Total	11 (61.1%)	4 (22.2%)	3 (16.7%)

(CR): platelet count of ≤400 x 10^9/L maintained for at least 4 weeks. Partial Response (PR): a platelet count of ≤600 x 10^9/L or a ≥50% reduction from baseline and maintenance of the reduction for at least 4 weeks. Nonresponse (NR): failure to meet CR or PR criteria.

Table 3. Related TEAEs

All Subjects*

	G1/2	G3	G4	Any		G1/2	G3	G4	Any
GI Symptoms	14	1	0	15	**Respiratory, Thoracic and Mediastinal**	2	0	0	2
Nausea	3			3	Dyspnoea	2			2
Abdominal pain	3			3	**Musculoskeletal and Connective Tissue**	1	0	0	1
Abdominal discomfort	1			1	Bone Pain	1			1
Diarrhea	4			4	**Hepatobiliary**	1	0	1	2
Abdominal bloating	2			2	Drug-induced liver injury	1			1
Acute pancreatitis		1		1	Cholestatic jaundice			1	1
Vomiting	1			1	**Blood and Lymphatic**	1	1	0	2
General Disorders	6	0	0	6	Anemia		1		1
Fatigue	3			3	Thrombocytopenia	1			1
Malaise	1			1	**Skin and Subcutaneous Tissue**	2	0	0	2
Edema peripheral	2			2	Night sweats	1			1
Nervous System	8	2	0	10	Pruritus	1			1
Headache	6	2		8	**Investigations**	4	1	0	5
Dizziness	1			1	AST increased	1			1
Dysgeusia	1			1	ALT increased	1			1
Cardiac Disorder	5	0	0	5	Blood alkaline phosphate increased	1			1
Palpitations	4			4	Blood creatinine increased	0	1		1
Tachycardia	1			1	Heart rate increased	1			1
Vascular	2	0	0	2					
Hypertension	1			1					
Hypotension	1			1					

*Fourteen of eighteen subjects enrolled experienced a treatment related adverse event

Table 4. Study Discontinuations

Rationale	n	%
Toxicity / Intolerance	5	27.8
Investigator Decision	2	11.1
Subject Request	2	11.1

Table 5. Pharmacokinetics: GALE-401 MPN and HV

			Subjects	Healthy Volunteers	
	Study		**GALE-401-201**	**BIO-ANA104 + BIO-ANA105**	
	N		18	38	
	Diagnosis		MPN (72.0% ET)		
	Age [years]	Mean	62.8	32.6	
	Treatment		**0.5 mg CR**	**0.5 mg CR**	**0.5 mg IR**
Parent	Cmax [pg/mL]	Geometric Mean (Mean)	1410.2 (1687)	526.3 (702.9)	2440.3 (2781.9)
	Tmax [hrs]	Geometric Mean (Mean)	2.2 (2.3)	2.3 (2.5)	1.0 (1.1)
	Terminal T1/2 [hrs]	Mean	20.2*	19.6	1.6
3-OH	Cmax [pg/mL]	Geometric Mean (Mean)	2347.8 (2639.8)	757.3 (853.1)	3435.5 (3646.5)
	Tmax [hrs]	Geometric Mean (Mean)	2.4 (2.5)	2.5 (2.8)	0.96 (1.1)
	Terminal T1/2 [hrs]	Mean	20.2*	17.2	3.0

* Based on population PK modeling, pooling MPN and HV data to predict elimination

Figure 3. PK Plasma Concentration / Time

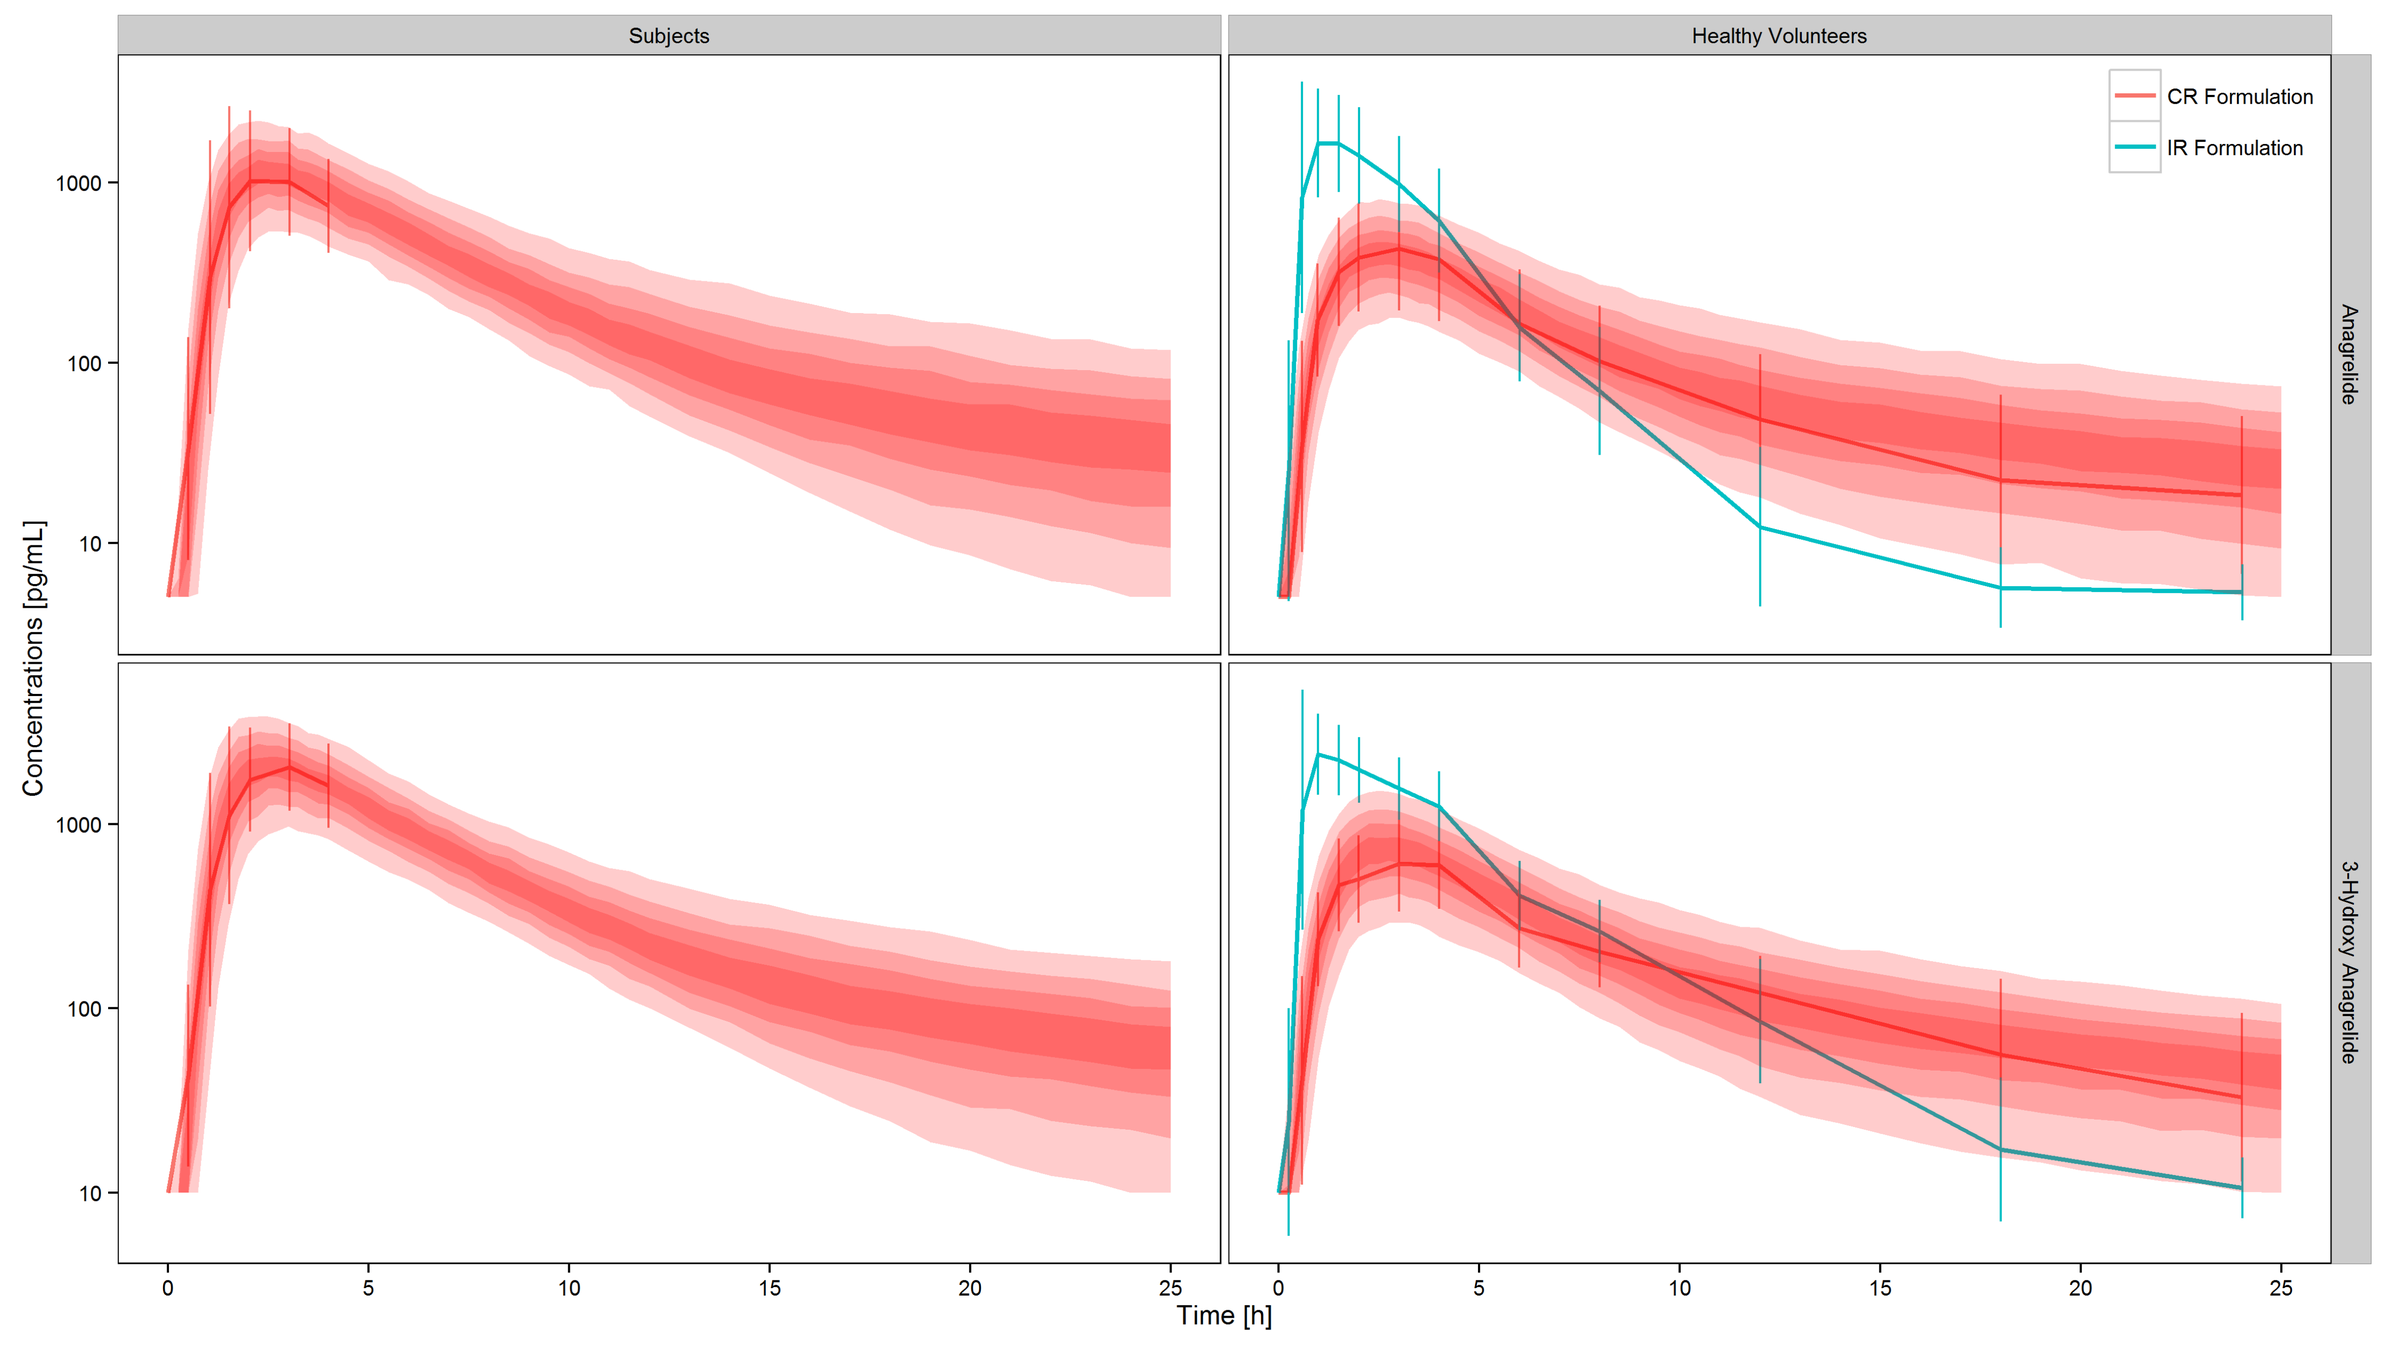
Subjects
Healthy Volunteers
CR Formulation
IR Formulation
Anagrelide
3-Hydroxy Anagrelide
Concentrations [pg/mL]
Time [h]
10
100
1000
0
5
10
15
20
25

The red shaded bands represent the distribution percentiles, from 10% to 90% by increments of 10%, of the predicted concentrations. The solid lines and the error bars represent the mean and the standard deviation of the observed concentrations. A 2-compartment model with first order absorption, time-varying coefficient of absorption, linear distribution and linear elimination (NONMEM 7.3 Icon Development Solutions) was suitable to characterize the PK of GALE-401 and its metabolite. The HV data was pooled with the MPN subject data to account for the lack of time-points after 4h.

Conclusions

- ◆ Study GALE-401-201 demonstrated GALE-401 is well tolerated in the number of subjects evaluated.
- ◆ GALE-401 efficacy compares favorably to historical anagrelide IR (Harrison et al. 2005; Gisslinger et al. 2013) response rates: ORR (70-80%), CR (50-60%), PR (10-20%).
- ◆ Compared to historical anagrelide IR (product monograph), the mean time to response, defined as PLT count ≤ 600 x10^9/L ranged from 4–12 wks compared to 1–9 wks observed with GALE-401 in MPN subjects.
- ◆ Population PK and non-compartmental analysis of PK parameters demonstrated a 1.5-fold and 3-fold respectively increased exposure of parent and 3-OH anagrelide metabolite compared to healthy volunteers (Laliberte R et al. 2014). Disease status, age, gastric emptying time and metabolic efficiency may be associated with the observed differences in the populations.
- ◆ GALE-401 remains a viable potential treatment option for MPN subjects. A randomized trial comparing GALE-401 vs. anagrelide IR in anagrelide naïve subjects is warranted.

- ◆ Ahluwalia M, Donovan H, Singh N, et al. J Thromb Haemost. 2010;8(10):2252-61.
- ◆ Laliberte RJ, Glidden PF, and Hamilton BL. Abstract 3178. American Society of Hematology, 2014.
- ◆ Verstovsek S, Saltzman M, Dakhil S, et al. Abstract P671. European Hematology Association, 2015.
- ◆ Harrison CN, Cambell PJ, Buck G et al. N Engl J Med 2005; 353:33-45.
- ◆ Gisslinger H, Gotic M, Holowiecki J et al. Blood 2013; 121:1720-1728.

GALE-401-201 study is sponsored by Galena Biopharma, Inc. Contact Remy Bernarda at ClinicalTrials@galenabiopharma.com for permission to reprint and/or distribute.

Conflict of interest disclosures: Research funding from Galena - Truong P, Saltzman M, Bessudo A, Jawien W, Lyons R, Berenzon D, Mena R, Verstovsek S; employees of Galena – Wingate-Pearse N, Choy G S ; contract research organization for Galena – Beeson H, Barriere O; Galena consultancy, equity ownership and patents royalties – Glidden P.

GALENA BIOPHARMA

American Society of Hematology, 57th Annual Meeting and Exposition, Orlando, FL December 5–8, 2015